P5-05-09

Trastuzumab deruxtecan combined with pyrotinib in first-line HER2-positive unresectable or metastatic breast cancer: an exploratory, multi-center, single-arm, phase Ib/II study (TROPHY)

Ying Fan[1], Huihui Li[2], Huiping Li[3], Zhenchuan Song[4], Ling Xu[5], Jin Yang[6], Jing Yao[7], Haijun Yu[8], Danyang Ji[1], Yuhang Han[1], Binghe Xu[1]

[1]Department of Medical Oncology, National Cancer Center/National Clinical Research Center for Cancer/Cancer Hospital, Chinese Academy of Medical Sciences and Peking Union Medical College, Beijing, China. [2]Department of Medical Oncology, Shandong Cancer Hospital and Institute, Shandong First Medical University and Shandong Academy of Medical Sciences, Jinan, China. [3]Department of Breast Oncology, Peking University Cancer Hospital and Institute, Beijing, China. [4]Breast Center, Fourth Hospital of Hebei Medical University, Shijiazhuang, China. [5]Department of Thyroid and Breast Surgery, Peking University First Hospital, Beijing, China. [6]Department of Medical Oncology, The First Affiliated Hospital of Xi'an Jiaotong University, Xi'an, China. [7]Cancer Center, Union Hospital, Tongji Medical College, Huazhong University of Science and Technology, Wuhan, China. [8]Department of Radiation and Medical Oncology, Zhongnan Hosital of Wuhan University, Wuhan, China.

San Antonio Breast Cancer Symposium
December 10-13, 2024

Background

- Trastuzumab deruxtecan (T-DXd), a human epidermal growth factor receptor 2 (HER2)-directed antibody-drug conjugate (ADC), has demonstrated remarkable efficacy in the second-line or later treatment of HER2-positive (HER2+) metastatic breast cancer (mBC) in DESTINY-Breast03 [1-2], and its exploration as the first-line (1L) treatment (DESTINY-Breast09) is ongoing[3].
- Pyrotinib, an irreversible, pan-HER receptor tyrosine kinase inhibitor (TKI)[4], has also been approved in combination with trastuzumab and docetaxel for the 1L treatment of HER2+ mBC and is widely used in China[5].
- As preclinical and clinical studies have shown the synergistic effects of HER2 ADCs combined with TKIs[6-7], this phase Ib/II study evaluates the efficacy and safety of the promising combination of T-DXd and pyrotinib for the 1L treatment of HER2+ mBC in China (TROPHY, NCT06245824).

Methods

- This multi-center, single-arm, phase Ib/II study was conducted in 8 sites in China and is comprised of a dose finding stage (phase Ib) followed by a dose expansion stage (phase II).
- Patients with no prior chemotherapy or HER2-targeted therapy in the metastatic setting who have experienced relapse with a disease-free interval (DFI) > 6 months from the completion of neoadjuvant or adjuvant treatment to advanced or metastatic diagnosis are eligible for enrollment. Asymptomatic or treated brain metastases is allowed.
- The primary objective of phase Ib was to assess safety and tolerability and determine the recommended phase 2 dose (RP2D). Patients were followed up beyond the 21-day dose-limiting toxicity (DLT) period for safety events.
- The primary objective of phase II was to evaluate the efficacy of T-DXd plus pyrotinib as the 1L treatment of HER2+ mBC.

Study Design

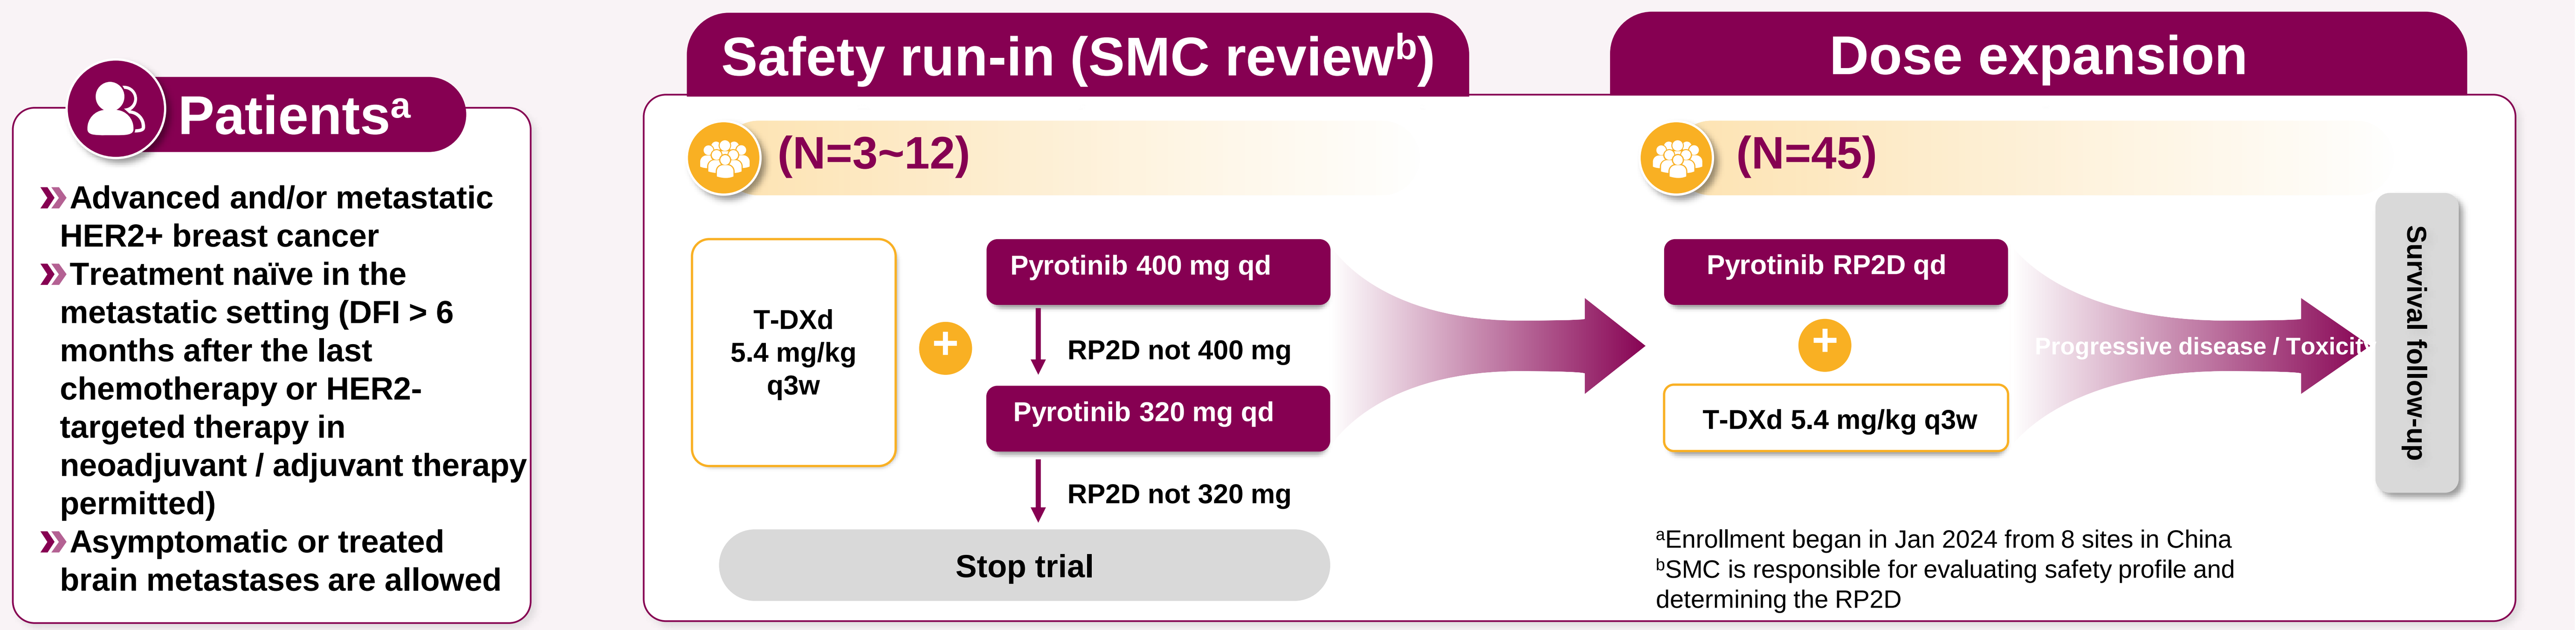
Patients[a]
Advanced and/or metastatic HER2+ breast cancer
Treatment naïve in the metastatic setting (DFI > 6 months after the last chemotherapy or HER2-targeted therapy in neoadjuvant / adjuvant therapy permitted)
Asymptomatic or treated brain metastases are allowed
Safety run-in (SMC review[b])
(N=3~12)
T-DXd 5.4 mg/kg q3w
Pyrotinib 400 mg qd
RP2D not 400 mg
Pyrotinib 320 mg qd
RP2D not 320 mg
Stop trial
Dose expansion
(N=45)
Pyrotinib RP2D qd
T-DXd 5.4 mg/kg q3w
Progressive disease / Toxicity
Survival follow-up

[a]Enrollment began in Jan 2024 from 8 sites in China
[b]SMC is responsible for evaluating safety profile and determining the RP2D

Results

- As of 28 Jun 2024, 5 HER2+ mBC patients were treated with T-DXd 5.4 mg/kg every 21 days (Q3W) plus pyrotinib 400 mg (n=2) or 320 mg (n=3) once daily as starting dose, with a median duration of follow up of 3.5 months (range, 0.7-5.4). The median age of enrolled patients was 47 years (range, 33-68), and all of them had an ECOG performance status (PS) of 1, 60% (3 of 5) with visceral metastases (Table 1).
- The only DLT was grade 3 anorexia, which occurred during the first cycle in 1 patient who received 400 mg of pyrotinib. This was followed by a de-escalation to the pyrotinib 320 mg cohort, where no DLT occurred.
- The most common treatment-emergent adverse events (TEAEs) included diarrhea (100%, 5 of 5), nausea (100%, 5 of 5), vomiting (100%, 5 of 5), anorexia (60%, 3 of 5), alanine aminotransferase (ALT) increased (60%, 3 of 5), aspartate aminotransferase (AST) increased (60%, 3 of 5), and weight loss (60%, 3 of 5), etc. Grade 3 TEAEs included anorexia and ALT increased (in 1 patient in the pyrotinib 400 mg cohort), and diarrhea (in 1 patient in the pyrotinib 320 mg cohort). There was no grade 4 or 5 TEAEs (Table 2).
- All 4 patients whose tumors were evaluable had achieved partial responses.

Conclusions

Pyrotinib 320 mg was identified as RP2D. The combination regimen of T-DXd plus pyrotinib showed a manageable safety profile with preliminary antitumor activity as the 1L treatment in patients with HER2+ mBC and will be explored in phase II stage.

Table 1. Baseline characteristics

		Pyrotinib 400 mg (n=2)	Pyrotinib 320 mg (n=3)	Total (n=5)
Age, years - mean (range)		56 (47-65)	48 (33-68)	51 (33-68)
Female sex - no.(%)		2 (100)	3 (100)	5 (100)
ECOG PS - no.(%)				
	0	0	0	0
	1	2 (100)	3 (100)	5 (100)
HR Status - no.(%)				
	Positive	0	2 (66.7)	2 (40)
	Negative	2 (100)	1 (33.3)	3 (60)
HER2 Status - no.(%)				
	IHC 3+	2 (100)	2 (66.7)	4 (80)
	IHC 2+, ISH+	0	1 (33.3)	1 (20)
Sum of diametersof target lesions, mm - mean (range)		20 (18-22)	93 (36-138)	63.8 (18-138)
Metastasis - no.(%)				
	Liver	2 (100)	1 (33.3)	3 (60)
	Lung	0	1 (33.3)	1 (20)
	Brain	0	0	0
	Bone	2 (100)	1 (33.3)	3 (60)
	Others	0	3 (100)	3 (60)
Previous therapy in the context of (neo)adjuvant setting-no.(%)				
	Chemotherapy	1 (50)	2 (66.7)	3 (60)
	Anti-HER2 therapy	1 (50)	1 (33.3)	2 (40)

Table 2. TEAEs of all grades that occurred in all patients and of grade ≥3 that occurred in one or more patients from screening visit until data cut-off date

TEAEs	Pyrotinib 400mg (n=2)		Pyrotinib 320mg (n=3)		Total (n=5)	
	No.	%	No.	%	No.	%
Diarrhea	2	100	3	100	5	100
Grade ≥3	0		1	33.3	1	33.3
Nausea	2	100	3	100	5	100
Vomiting	2	100	3	100	5	100
Anorexia	1	50	2	66.7	3	60
Grade ≥3	1	50	0		1	20
ALT increased	2	100	1	33.3	3	60
Grade ≥3	1	50	0		1	20
AST increased	2	100	1	33.3	3	60
Weight loss	2	100	1	33.3	3	60
Creatinine increased	2	100	0		2	40
Dental ulcer	2	100	0		2	40
Anemia	1	50	1	33.3	2	40
WBC decreased	1	50	1	33.3	2	40
Hypokalemia	1	50	1	33.3	2	40
Constipation	1	50	1	33.3	2	40
Dizziness	0		2	66.7	2	40
Asthenia	0		2	66.7	2	40
Pain of skin	0		2	66.7	2	40
Thrombocytopenia	1	50	0		1	20
Hypoalbuminemia	1	50	0		1	20
Urine output decreased	1	50	0		1	20
Dyspepsia	1	50	0		1	20
Neutrophil count decreased	0		1	33.3	1	20
GGT increased	0		1	33.3	1	20
ALP increased	0		1	33.3	1	20
Headache	0		1	33.3	1	20
Abdominal pain	0		1	33.3	1	20
Hyperhidrosis	0		1	33.3	1	20
Fever	0		1	33.3	1	20
Somnolence	0		1	33.3	1	20
Alopecia	0		1	33.3	1	20

Abbreviations

SMC, safety monitoring committee;; ECOG, Eastern Cooperative Oncology Group, ALP, alkaline phosphatase; ALT, alanine aminotransferase; AST, aspartate aminotransferase; GGT, gamma-glutamyl transpeptidase; WBC, white blood cell

Contact

<Dr. Ying Fan>
<Department of Medical Oncology, National Cancer Center/National Clinical Research Center for Cancer/Cancer Hospital, Chinese Academy of Medical Sciences and Peking Union Medical College, Beijing, China>
Email: fanyingfy@medmail.com.cn

References

1. Ogitani Y, et al. Clin Cancer Res. 2016 Oct 15;22(20):5097-5108.
2. Hurvitz SA, et al. Lancet. 2023 Jan 14;401(10371):105-117.
3. Tolaney SM, et al. Annals of Oncology (2021) 32 (suppl_5): S457-S515.
4. Ma F, et al. J Clin Oncol. 2017 Sep 20;35(27):3105-3112.
5. Ma F, et al. BMJ. 2023 Oct 31;383:e076065.
6. Olson D, et al. Cancer Res Commun. 2023 Sep 25;3(9):1927-1939.
7. Borges VF, et al. JAMA Oncol. 2018 Sep 1;4(9):1214-1220..

Acknowledgments

We thank the patients who are participating in this study as well as their families and caregivers. This study is funded by AstraZeneca.

Disclosures

Dr. Ying Fan declares no conflicts of interest.

This presentation is the intellectual property of the author/presenter. Contact them at <<your email address>> for permission to reprint and/or distribute.

© Poster Template by Genigraphics® 1.800.790.4001 www.genigraphics.com